LBP-33

GELESIS Superabsorbent Hydrogel Prevents Hepatic Steatosis and Insulin Resistance in High Fat Diet–Induced NAFLD Pre-Clinical Model

A. Silvestri, MSc[1]; A. Sannino, PhD[2]; M. Vitale, PhD[1]; J. Mouries, PhD[1]; I. Spadoni, PhD[1]; C. Demitri, PhD[2]; E. Chiquette, PharmD[2]; M. Rescigno, PhD[1]

[1]**Humanitas Clinical and Research Center – IRCCS**, via Manzoni 56, 20089 Rozzano (Mi) – Italy; [2]**GELESIS Inc**, 501 Boylston, Boston, MA, USA

BACKGROUND

- Nonalcoholic fatty liver disease (NAFLD) is an emerging epidemic paralleling obesity and diabetes, without safe and effective clinical therapeutic options
- Obesity, insulin resistance, and disrupted gut homeostasis are major contributors to the pathophysiology of NAFLD and its progression to nonalcoholic steatohepatitis (NASH)
- GELESIS novel hydrogel platform technology is based on crosslinked, modified cellulose orally administered
- Gelesis100, the first in this platform technology, has demonstrated, in clinical studies, weight loss and improvement in insulin sensitivity
- Gel-B, with double the elasticity of Gelesis100, was designed to restore gut barrier integrity based on previous preclinical work

OBJECTIVES

Our aim was to evaluate whether Gel-B can prevent hepatic steatosis by restoring gut barrier function, reducing insulin resistance, affecting gut dysbiosis, and inducing weight loss, all mechanisms involved in NAFLD/NASH.

EXPERIMENTAL DESIGN

- C57BL/6J wild-type male mice were fed with either an isocaloric low-fat diet (LFD; 10% fat, 70% CHO) or high-fat diet (HFD; 45% fat, 36% CHO) for 18 weeks
- In parallel, 2 groups of mice were fed with HFD that was enhanced with low- or high-dose Gel-B

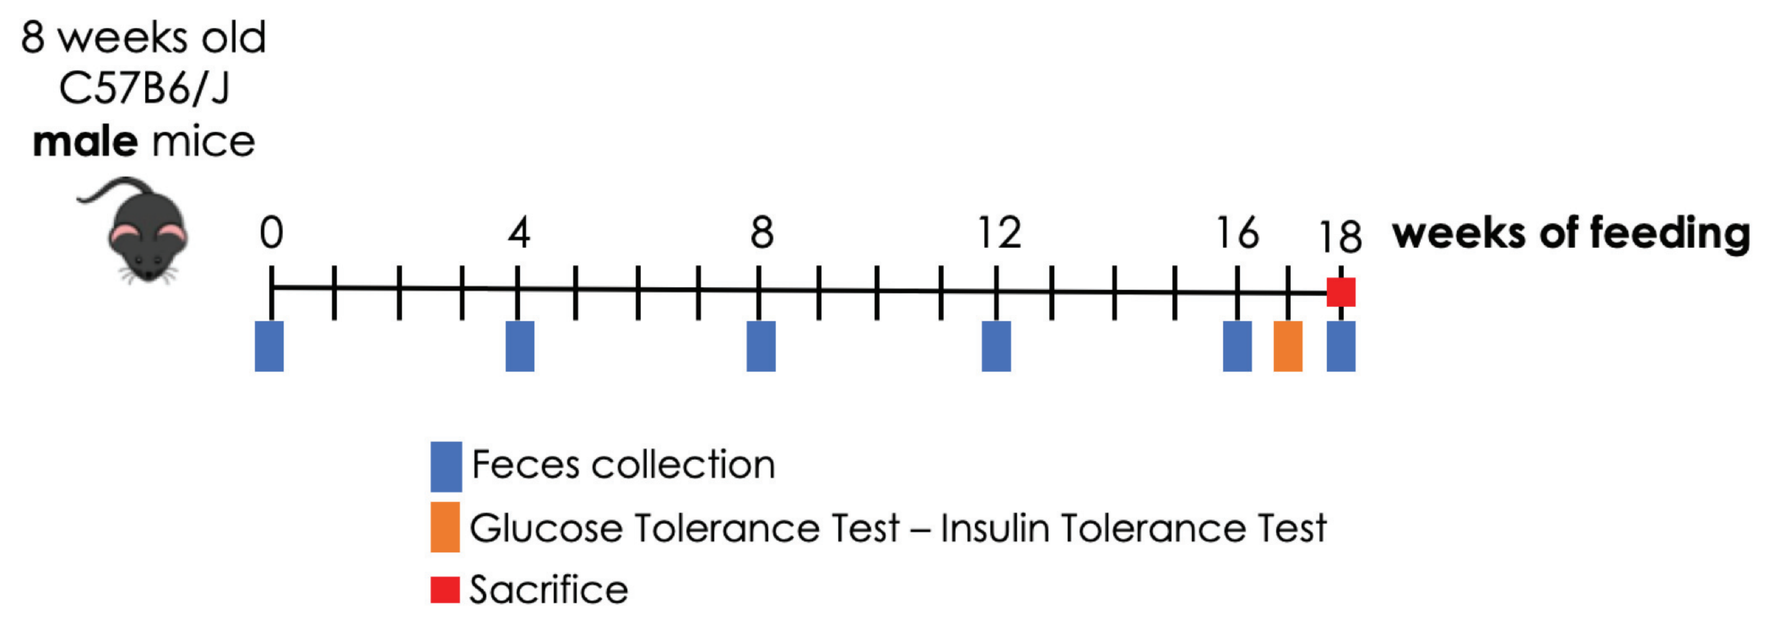
8 weeks old
C57B6/J
male mice
0
4
8
12
16
18
weeks of feeding
Feces collection
Glucose Tolerance Test – Insulin Tolerance Test
Sacrifice

- Changes in body weight, glucose/insulin tolerance, glucagon-like peptide-1 (GLP-1), and epidydimal adipose tissue (EAT) were measured
- Oil red O staining was performed to evaluate triglyceride accumulation in the liver
- Expression of zonula occludens-1 (ZO-1) and circulating levels of bacteria-derived lipolysaccharides (LPS) were evaluated to assess gut barrier function
- Feces were collected and total genomic DNA was extracted and sequenced

HUMANITAS RESEARCH HOSPITAL

RESULTS

Figure 1. Gel-B reduced body weight gain and EAT enlargement

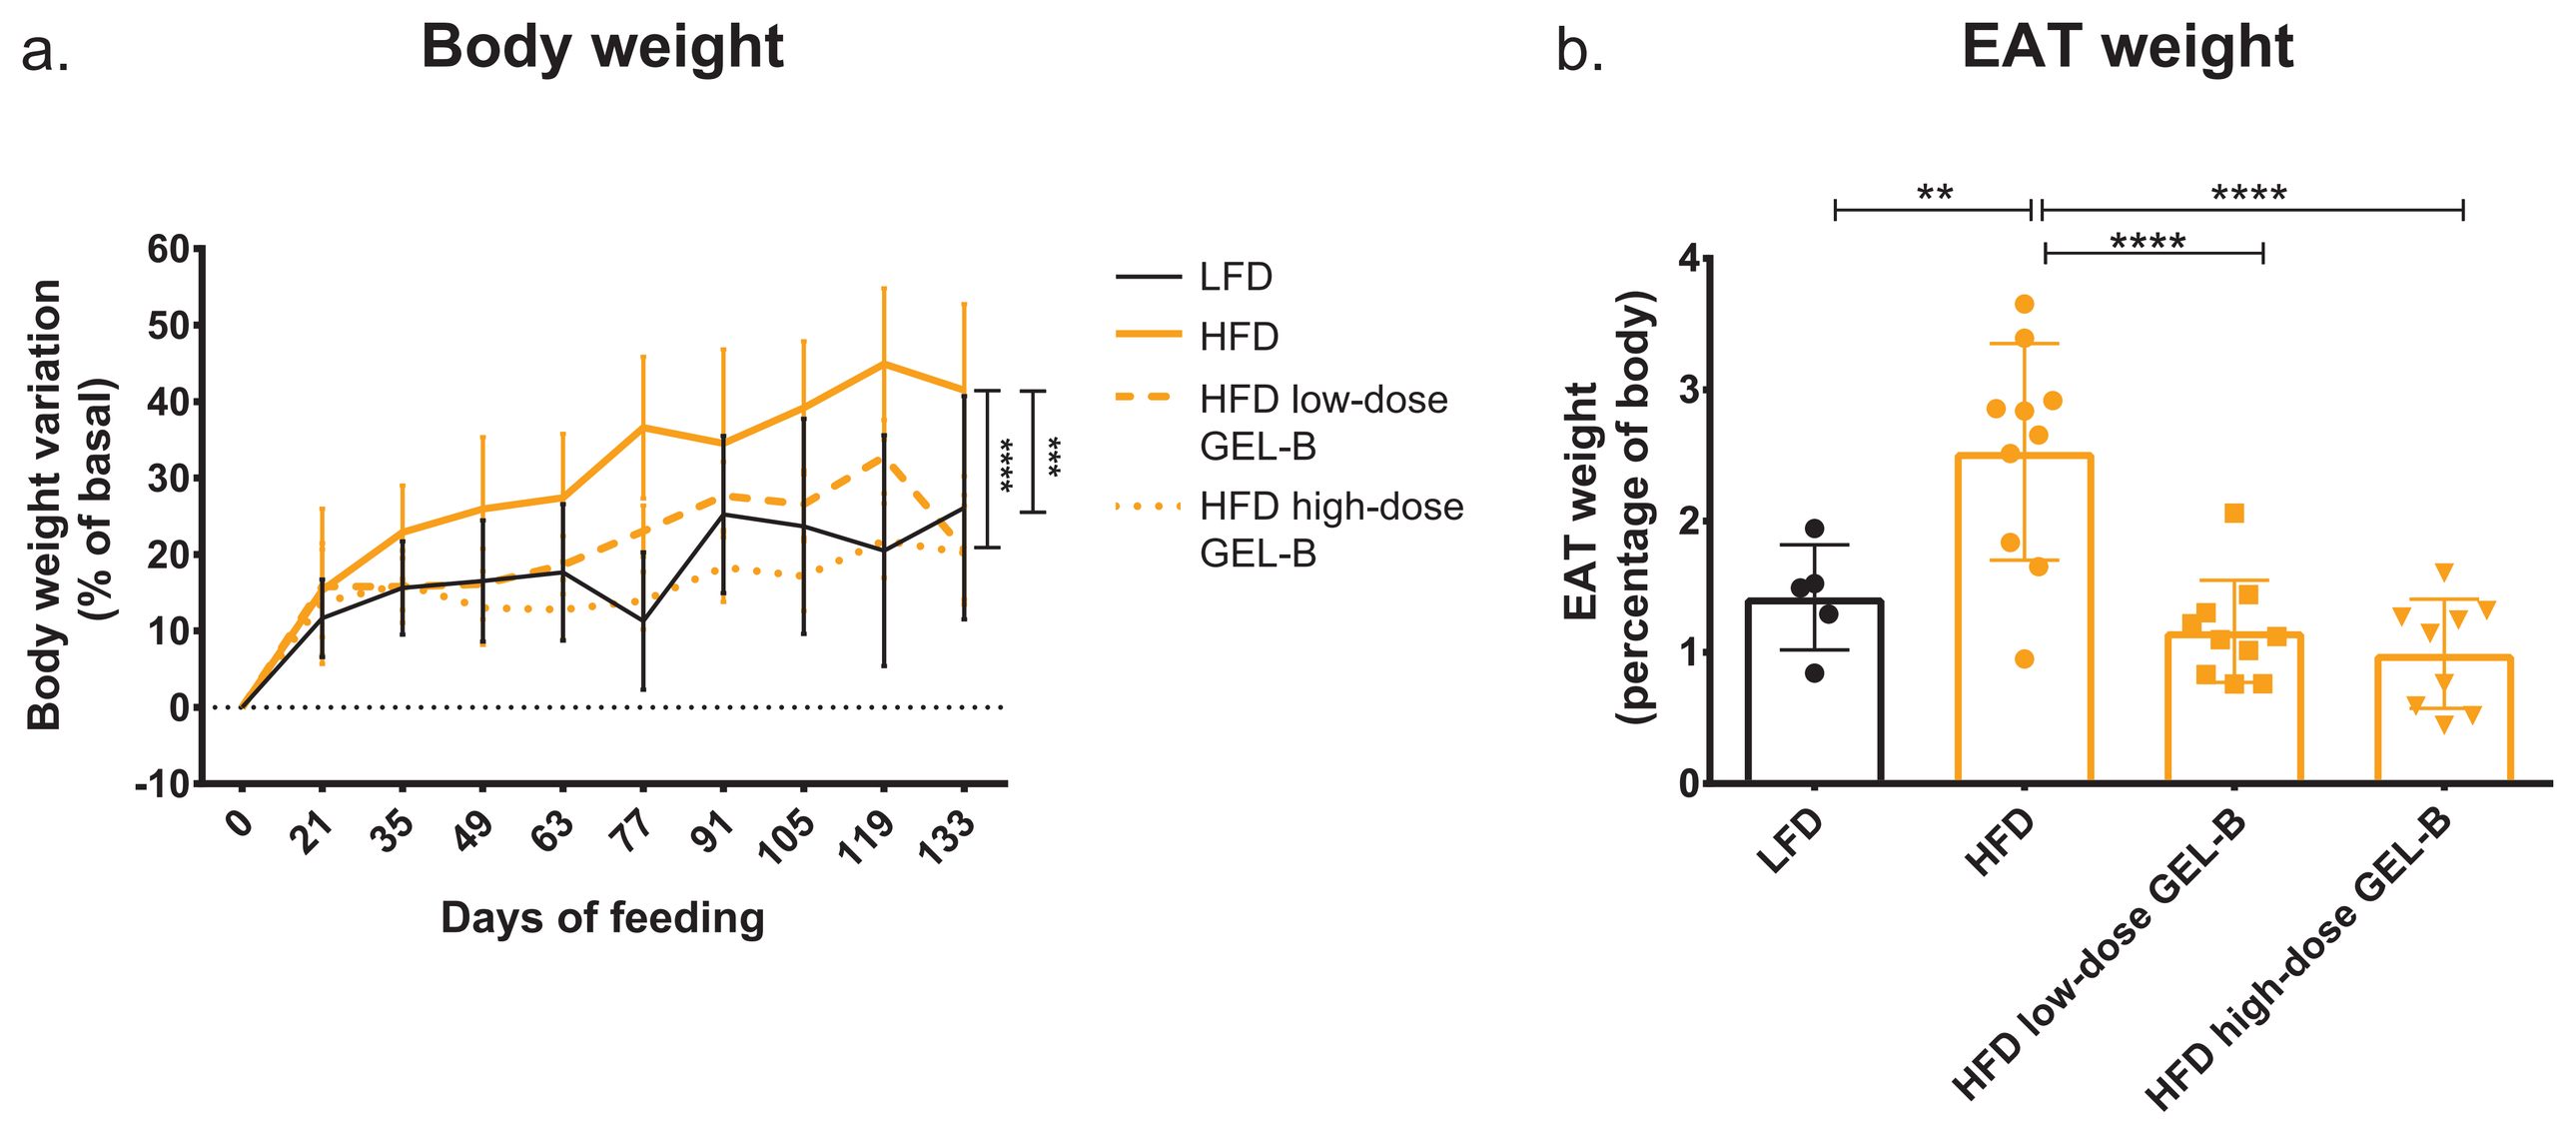
a.
Body weight
Body weight variation (% of basal)
Days of feeding
LFD
HFD
HFD low-dose GEL-B
HFD high-dose GEL-B
b.
EAT weight
EAT weight (percentage of body)
LFD
HFD
HFD low-dose GEL-B
HFD high-dose GEL-B

Body weight variation curves and epidydimal adipose tissue (EAT) weight at sacrifice.
a. Body weight curves expressed in percentage of basal, statistically significant difference in body weight between LFD vs HFD, HFD vs HFD low-dose Gel-B, and HFD vs HFD high-dose Gel-B (*****P* < 0.0001; two-way ANOVA; Bonferroni's multiple comparison test); b. EAT weight after 18 weeks of diet displayed in percentage of body weight (***P* <0.01; ****P* <0.001; one-way ANOVA; Bonferroni's multiple comparison test).

Gel-B reduced body weight gain to levels comparable to the low-fat diet and resulted in a strong reduction in EAT size, indicating that Gel-B administration was able to reduce white adipose tissue deposition.

Figure 2. Gel-B prevented triglyceride accumulation in liver

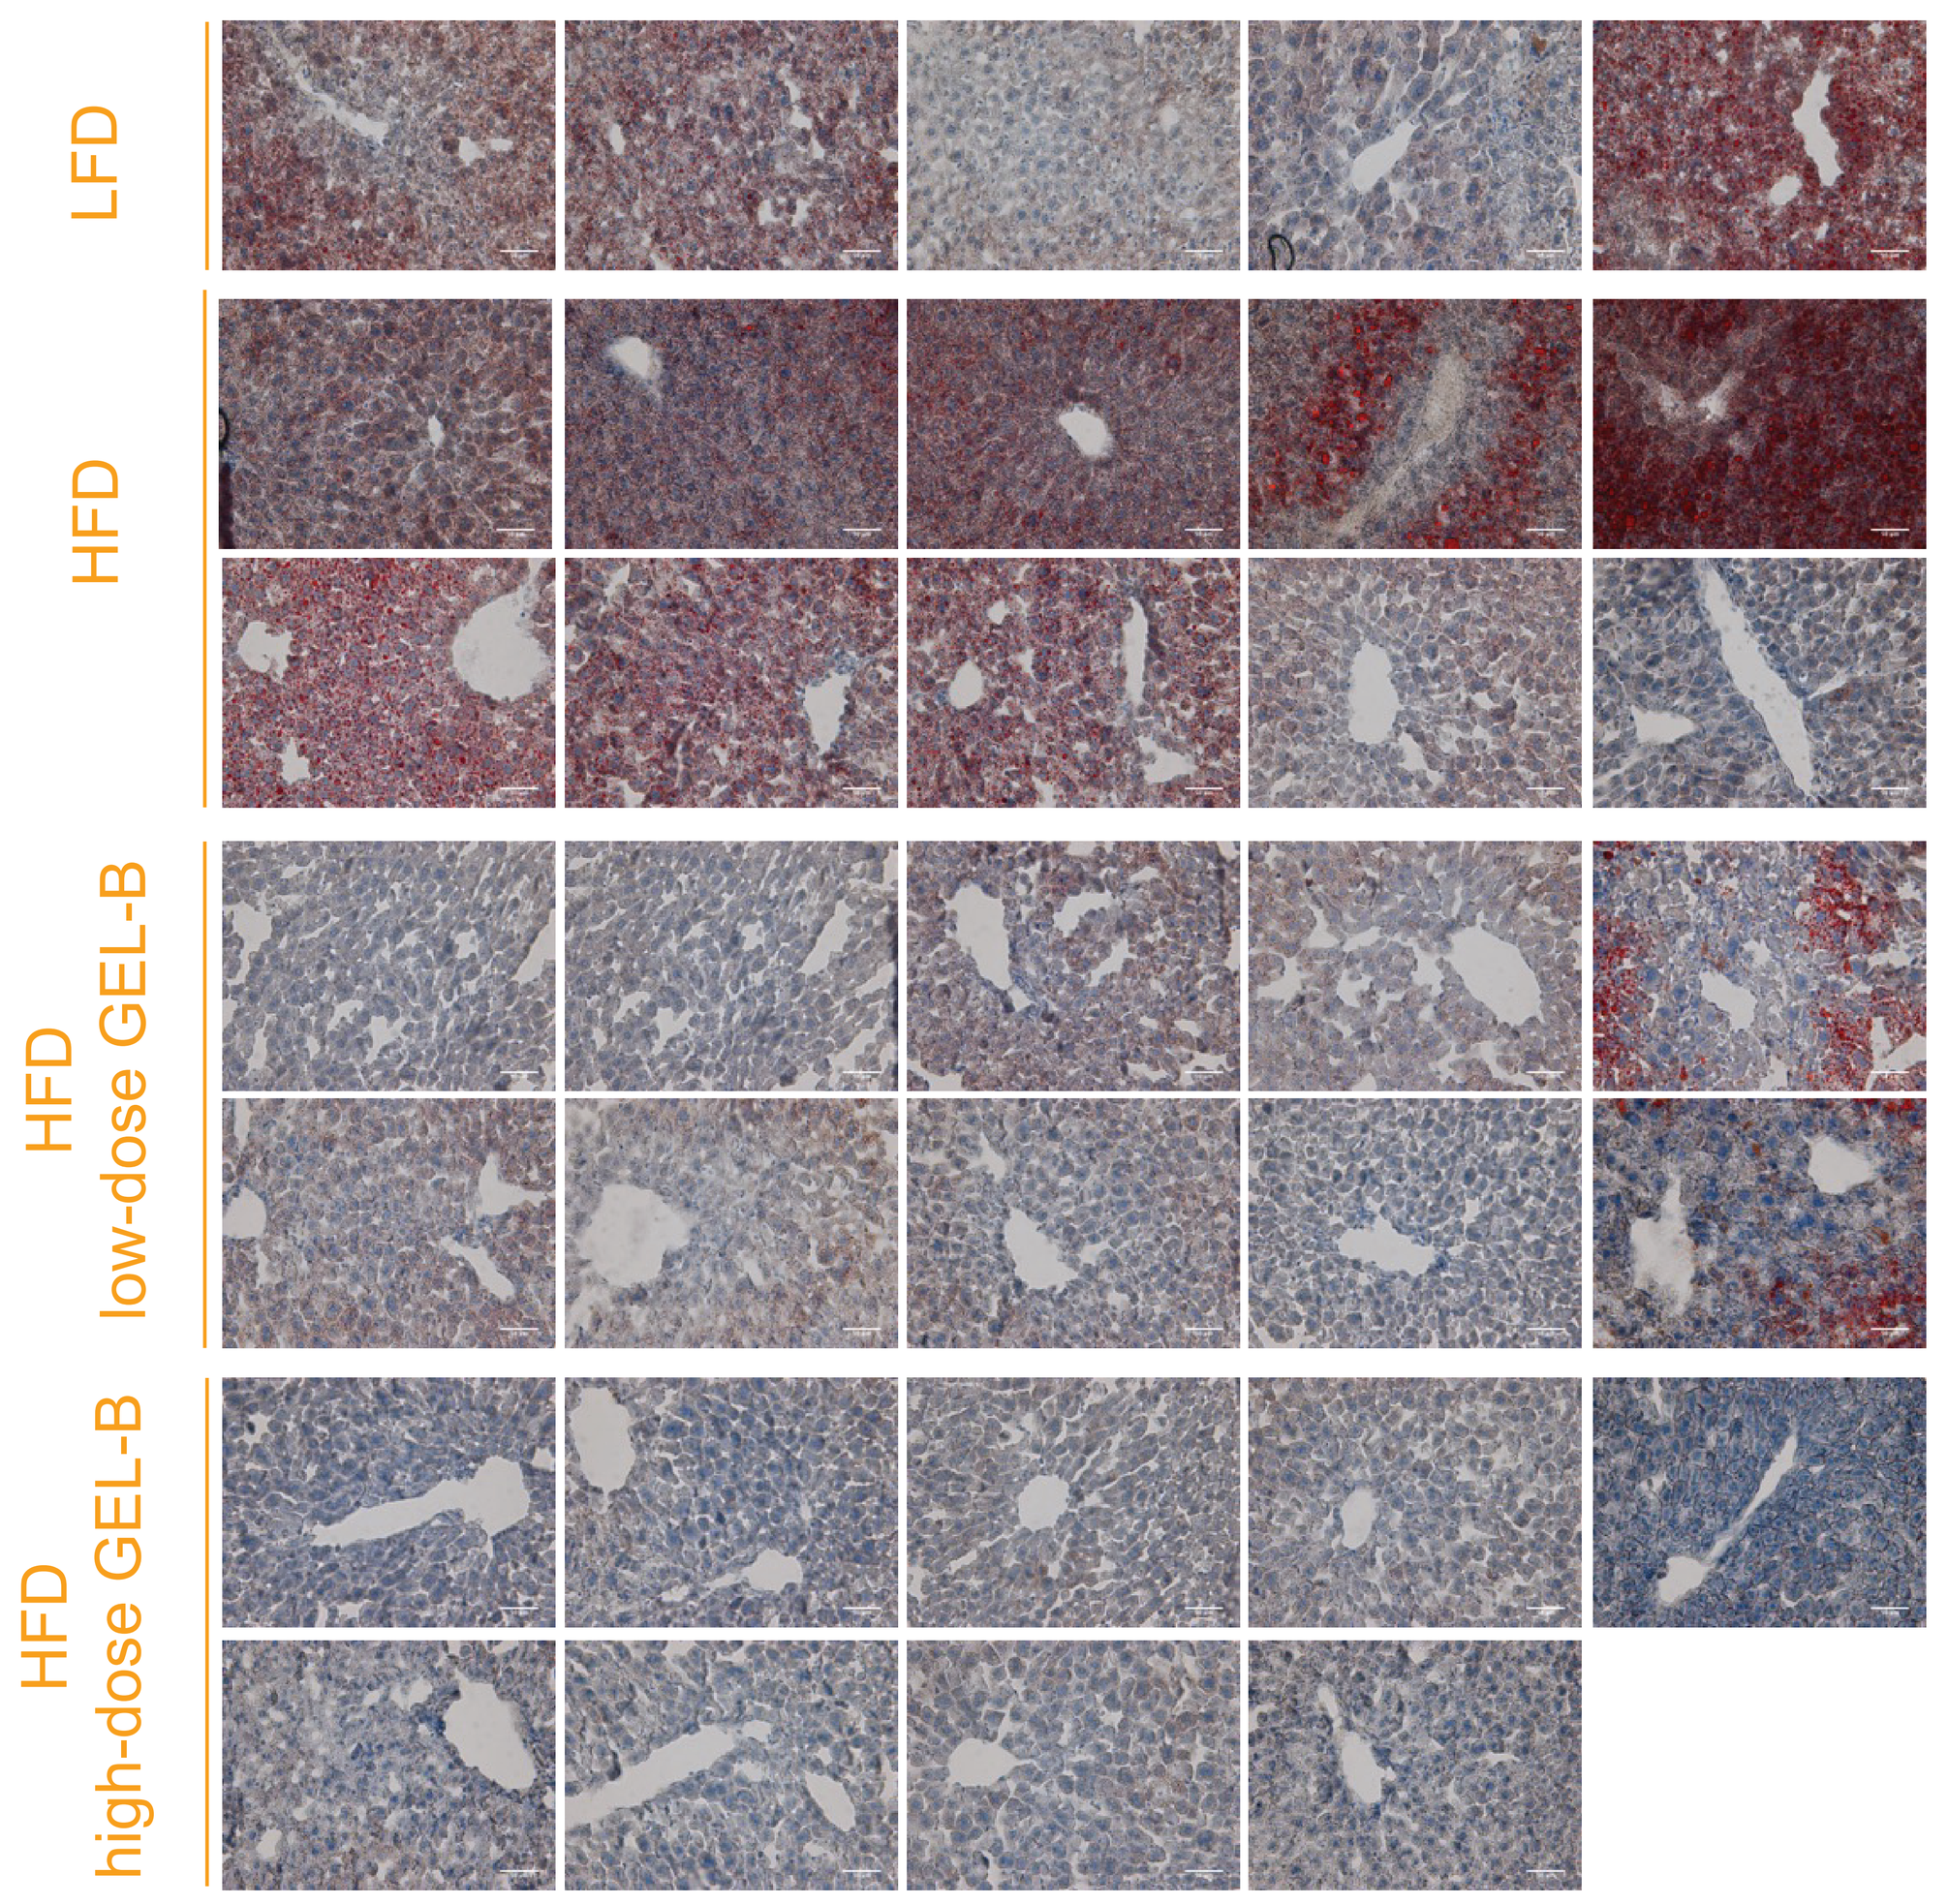
LFD
HFD
HFD low-dose GEL-B
HFD high-dose GEL-B

Liver triglyceride staining after 18 weeks of feeding. Oil Red O staining of liver sections, Oil Red O staining in red, hematoxylin (used as a counterstain for liver parenchyma) in blue; images from individual mice of each group, scale bar 50 µm.

Gel-B prevented liver triglyceride accumulation in a dose-dependant manner, indicating that Gel-B administration was protective against steatosis development in a model of high fat diet–induced obesity.

Figure 3. Gel-B prevented insulin-resistance development

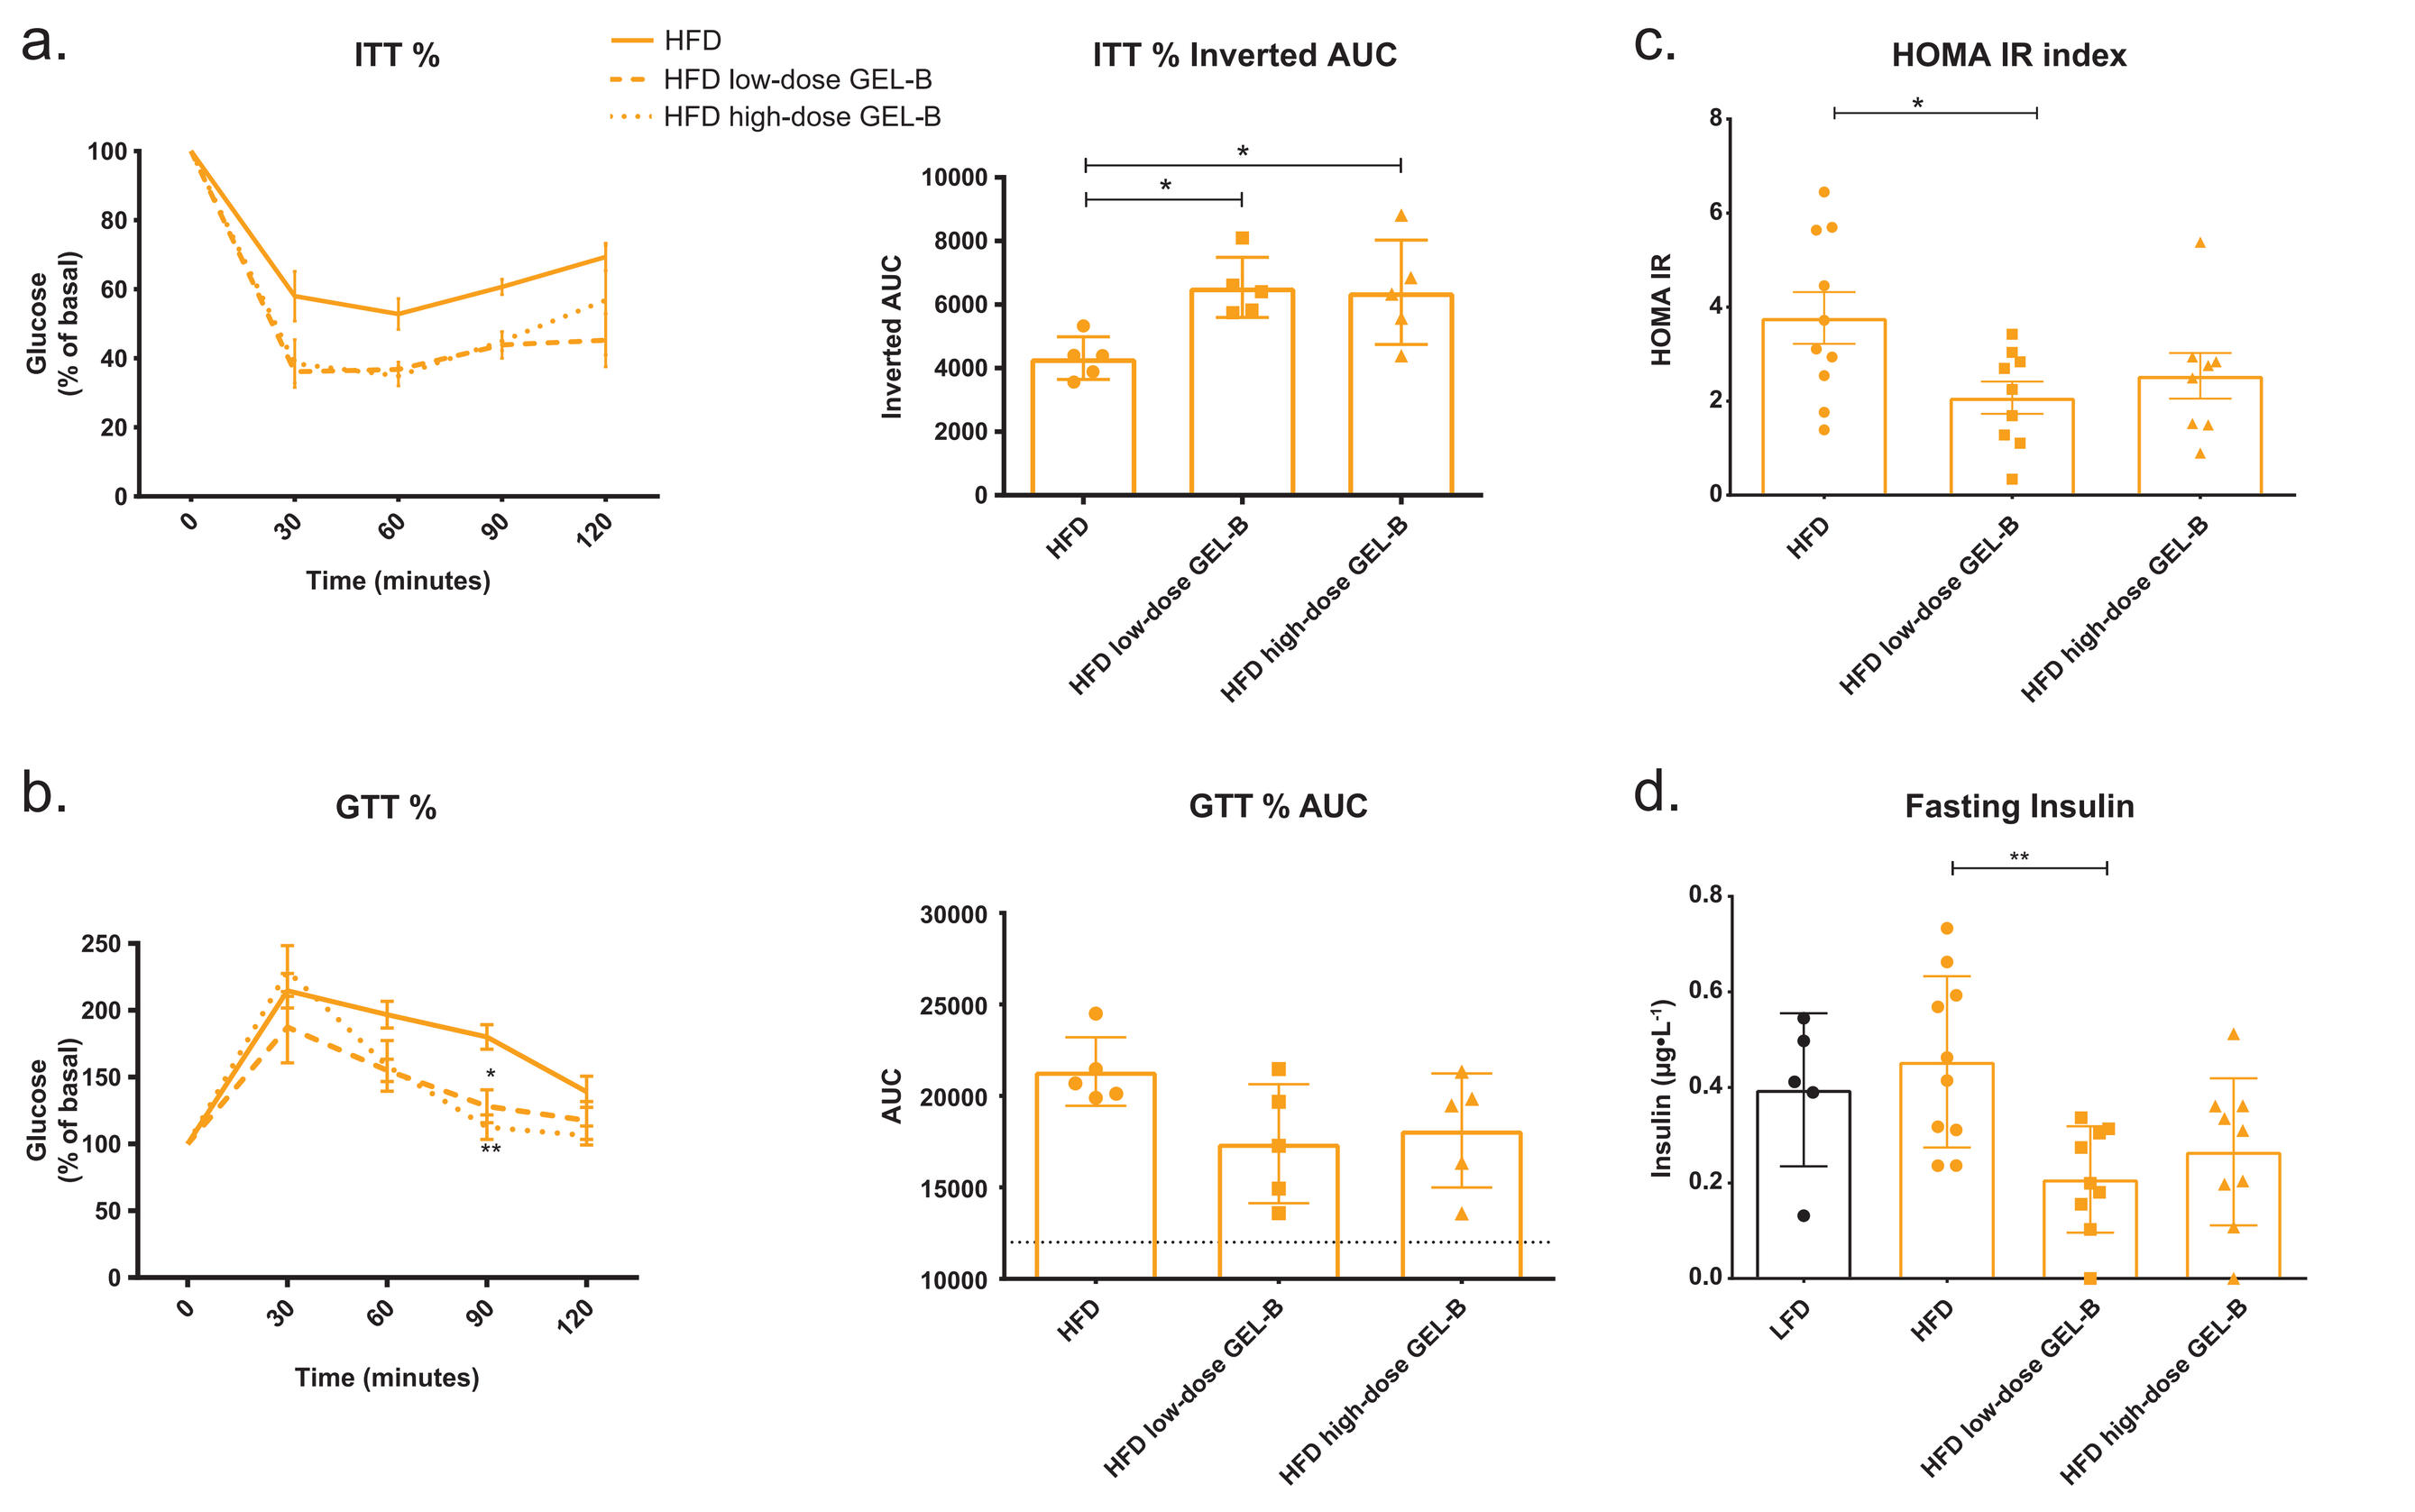
a.
ITT %
Glucose (% of basal)
Time (minutes)
HFD
HFD low-dose GEL-B
HFD high-dose GEL-B
ITT % Inverted AUC
Inverted AUC
c.
HOMA IR index
HOMA IR
b.
GTT %
Glucose (% of basal)
Time (minutes)
GTT % AUC
AUC
d.
Fasting Insulin
Insulin (µg•L⁻¹)

Insulin Tolerance test (ITT), Glucose Tolerance test (GTT), HOMA IR, and Glucagon-like peptide 1 (GLP-1).
a. Intra-peritoneal ITT and respective inverted area under the curve (AUC); **b.** Intra-peritoneal GTT and respective AUC. Both tests were performed after 17 weeks of feeding following 4 hours of morning fasting; **c.** HOMA IR values; **d.** Serum insulin levels measured after morning fasting, outliers were calculated with Graphpad Prism outlier calculator software and removed from the analysis (**P* <0.05, ***P* <0.01, one-way ANOVA, Bonferroni's multiple comparisons test).

Gel-B treatment ameliorated response to insulin and glucose tolerance tests and reduced homeostatic model assessment of insulin resistance (HOMA IR) index values, highlighting Gel-B ability to protect against insulin resistance development in a murine model.

Figure 4. Gel-B administration increased GLP-1 serum levels

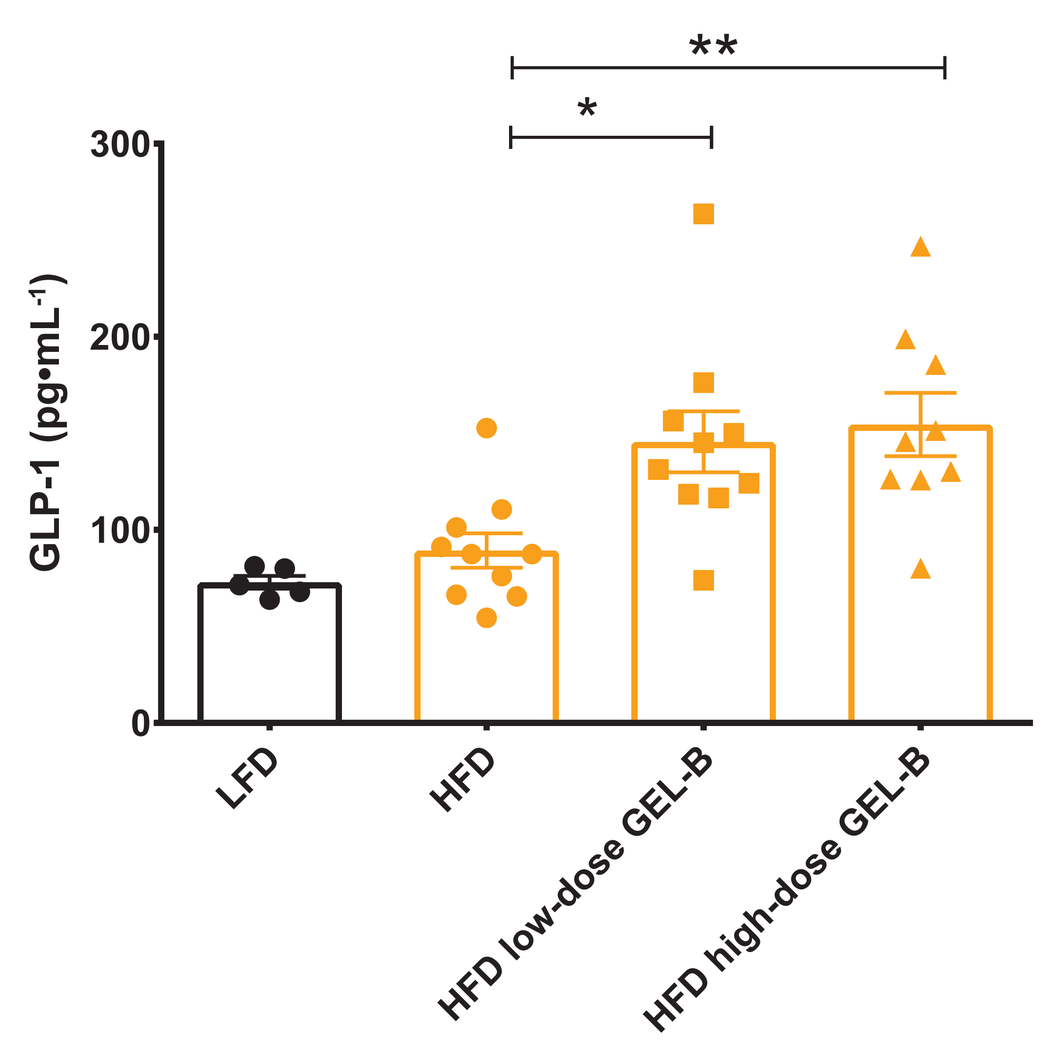
GLP-1 (pg•mL⁻¹)
LFD
HFD
HFD low-dose GEL-B
HFD high-dose GEL-B

Gel-B treatment induced a significant increase in serum GLP-1 levels.

GLP-1, a gut-derived peptide with known anti-diabetic and anti-steatotic effects.

Glucagon-like peptide 1 (GLP-1) serum levels after 18 weeks of feeding.
GLP-1 serum levels measured after morning fasting (**P* <0.05, ***P* <0.01, one-way ANOVA, Bonferroni's multiple comparisons test).

Figure 5. Gel-B administration modified gut microbiota composition

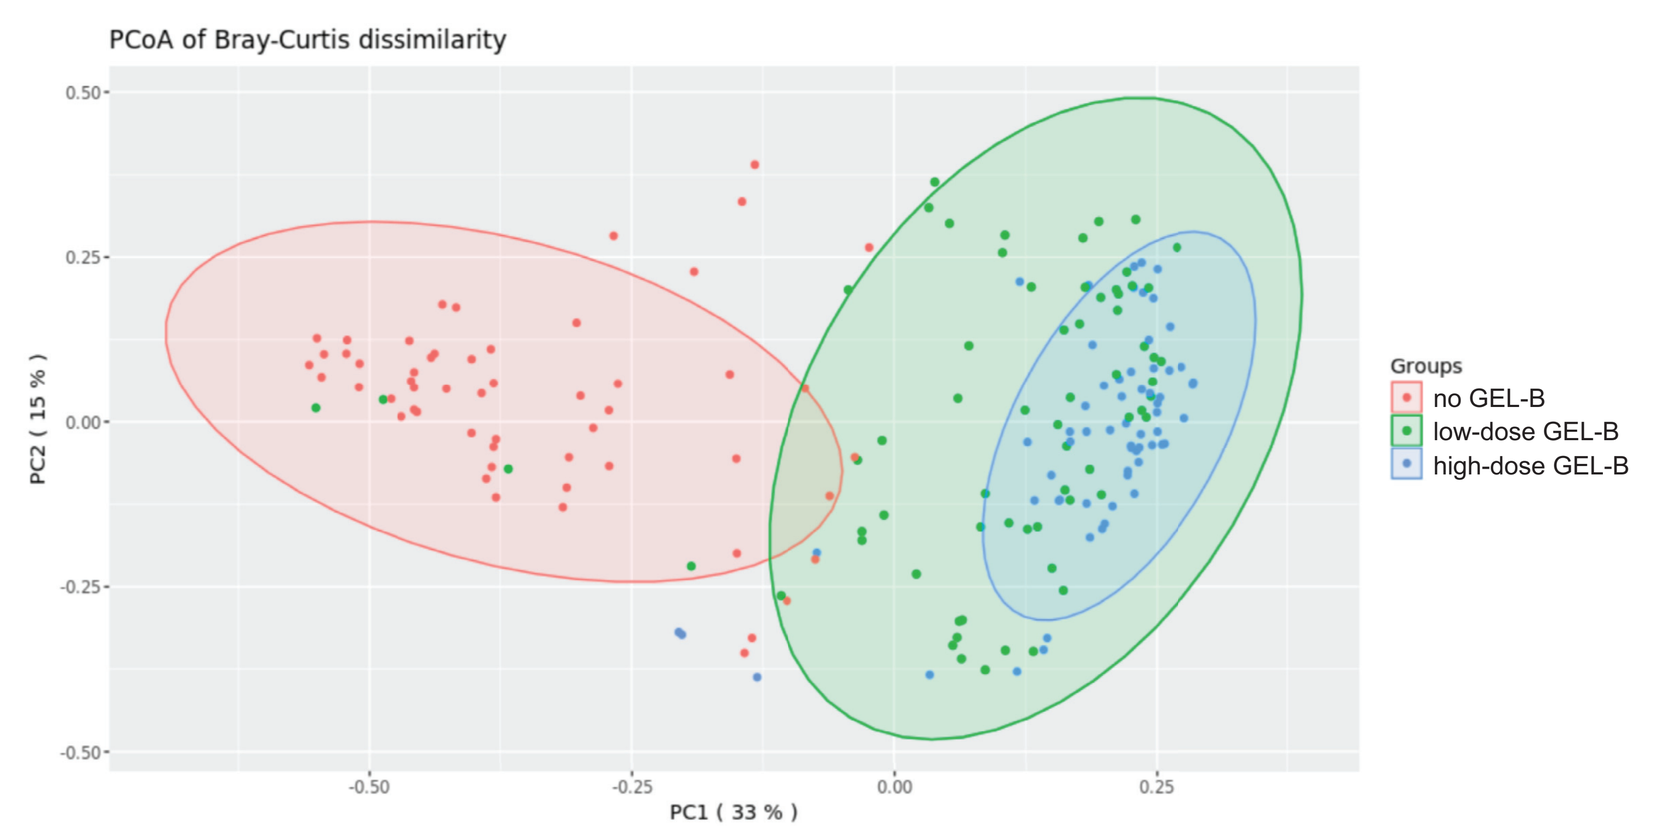
PCoA of Bray-Curtis dissimilarity
PC1 (33 %)
PC2 (15 %)
Groups
no GEL-B
low-dose GEL-B
high-dose GEL-B

Gel-B treatment is associated with a significant separation of microbial communities (P <0.001).

Principal Coordinates Analysis (PCoA) of Bray-Curtis dissimilarity for all samples. Points are colored by sample group, highlighting diet supplement. Red points indicate mice that received diet without Gel-B, in green mice that received lower dose of Gel-B, and in blue mice that received higher dose of Gel-B.

Figure 6. Gel-B improved gut barrier function

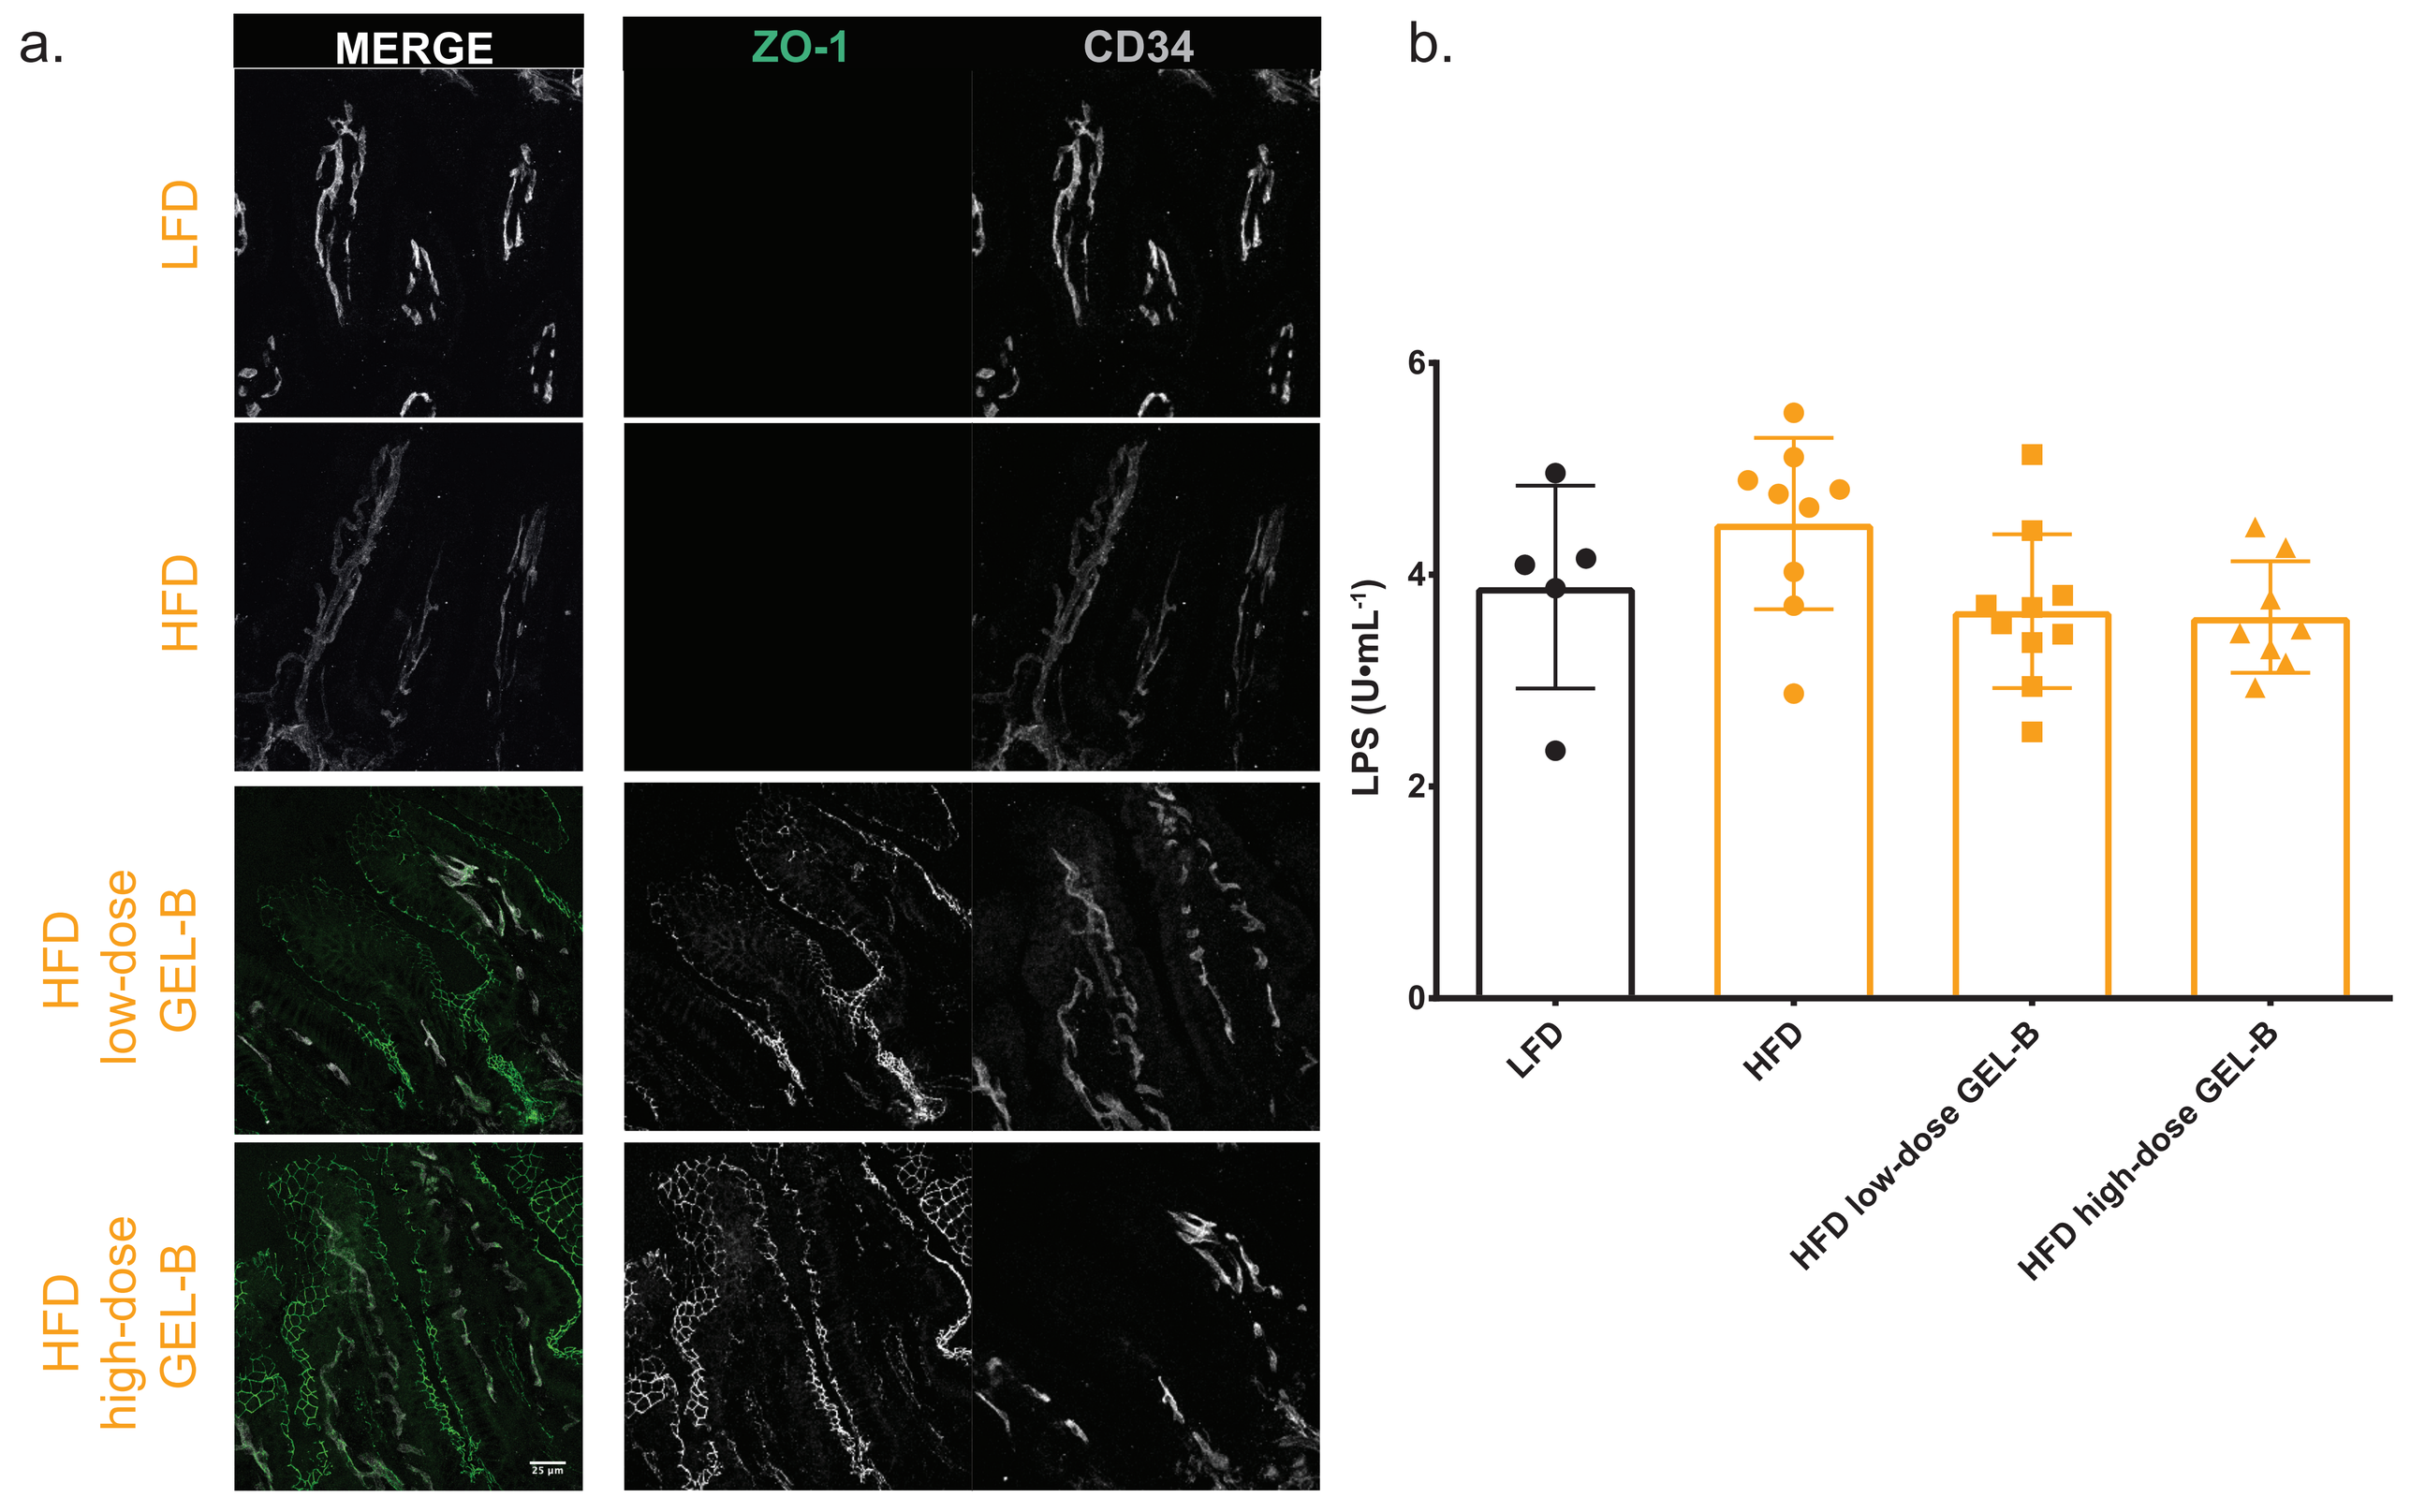
a.
MERGE
ZO-1
CD34
LFD
HFD
HFD low-dose GEL-B
HFD high-dose GEL-B
b.
LPS (U•mL⁻¹)
LFD
HFD
HFD low-dose GEL-B
HFD high-dose GEL-B

Evaluation of gut barrier function **a.** Ileum tissue samples were stained for zonula occludens (ZO-1) in green and CD34 (marker of endothelium) in grey. Representative images of n = 5 LFD group, n = 10 for HFD, HFD low-dose, and high-dose Gel-B. Scale bar 25 µm; **b.** Serum LPS levels after 18 weeks of feeding.

Gel-B treatment increased ZO-1 expression in gut epithelium and endothelium and limited translocation of LPS into blood, two indicators of improved gut barrier function.

CONCLUSIONS

Mice on HFD treated with Gel-B:

- Significantly decreased EAT and reduced weight gain
- Prevented hepatic steatosis (NAFLD)
- Significantly reduced HOMA IR, consistent with the improvement of glucose and insulin tolerance tests
- Increased total GLP-1 levels
- Increased ZO-1 expression and limited translocation of LPS into blood
- Induced a pronounced compositional separation of gut microbiota

These results support the role of Gel-B as a potential therapeutic option to target pathogenic mechanisms involved in NAFLD/NASH: excess weight, insulin resistance, gut barrier dysfunction, and gut dysbiosis.

Translating this research into the clinic could uncover a new therapeutic approach for this emerging epidemic.

DISCLOSURES

A. Silvestri: None. **A. Sannino:** Employee; Self; Gelesis. **M. Vitale:** None. **J. Mouries:** None. **I. Spadoni:** None. **C. Demitri:** Employee; Self; Gelesis. **E. Chiquette:** Employee; Self; Gelesis. **M. Rescigno:** None.

GELESIS

Presented at EASL International Liver Congress 2019, Vienna, Austria, 10 April 2019