Short-Term Administration of Gelesis200 Increases Fullness and Satiety in Overweight and Obese Subjects: First-in-Human Safety Study

Lorien E. Urban[1], Denis Audet[2], Eyal S. Ron[1], Alessandro Sannino[1], Yishai Zohar[1] and Hassan M. Heshmati[1]

[1]Gelesis, Boston, MA. [2]InVentiv Health Clinique, Québec, Canada.

ABSTRACT

Background:

- Gelesis100, a novel biocompatible hydrogel, increases satiety, reduces food intake, and induces weight loss when taken 30 min before meals. Gelesis200, an additional hydrogel in the product family with a higher elastic modulus and a faster hydration rate, is being developed for weight loss and glycemic control, particularly in subjects with prediabetes and type 2 diabetes.

Methods:

- Gelesis200 (2.10 g) or placebo capsules were administered to 24 overweight and obese male subjects 10 or 30 min before standard meals on a single day, in a double-blind, crossover fashion. Appetite was assessed using visual analogue scales (VAS). All subjects completed VAS after breakfast and lunch and 12 subjects also completed VAS after dinner. Results are differences in incremental area under the curve and individual timepoints.

Results:

- Gelesis200 was safe and well tolerated. Compared to placebo, administration of Gelesis200 at -10 min resulted in greater fullness after lunch (180 ± 398 cm*min; P = 0.046). The 12 subjects who completed all 3 meals had greater fullness during the entire day (704 ± 839 cm*min; P = 0.012). Administration of Gelesis200 at -10 min resulted in greater satiety just before dinner (2.3 ± 2.8 cm; P = 0.017) and 150 min (1.4 ± 2.1 cm; P = 0.032) and 180 min (1.5 ± 2.0 cm; P = 0.031) after dinner. When administered at -30 min, the above differences with placebo were not observed. Compared to administration at -30 min, administration at -10 min produced more fullness 90 min after lunch (1.0 ± 2.5 cm; P = 0.050) and less desire to eat 30 min (-0.8 ± 1.7 cm; P = 0.037), 90 min (-1.3 ± 2.0 cm; P = 0.005), and 120 min (-1.0 ± 2.1 cm; P = 0.039) after lunch, consistent with faster hydration kinetics of Gelesis200.

Conclusions:

- Short-term administration of Gelesis200 to overweight and obese subjects was safe and well tolerated. Administration 10 min before meals increased fullness and satiety, supporting the utility of Gelesis200, a rapidly acting hydrogel, in the management of obesity.

BACKGROUND

- Obesity is a major predisposing factor for prediabetes, type 2 diabetes, and other comorbidities. The worldwide prevalence of obesity has nearly doubled between 1980 and 2014 according to World Health Organization estimates.[1]
- Concomitant with this rise in obesity, the number of adults with diabetes worldwide has quadrupled since 1980.[2] In fact, 85% of people with type 2 diabetes are overweight or obese.[3]
- Weight loss of 5-10% lowers the risk of diabetic- and obesity-related comorbidities;[4] however, adoption of antiobesity pharmacotherapies is extremely low, suggesting systemic barriers to prescription.[5] One barrier is safety concerns based on a history of FDA withdrawals. This highlights an unmet need for safe and effective therapies to facilitate reduction of excess body weight.
- Gelesis100 is a novel, oral, non-systemic hydrogel being developed for weight loss in patients who are overweight and obese, with or without type 2 diabetes. Gelesis100 increases feelings of satiety, and reduces feelings of hunger,[6] and facilitates significant loss of body weight, particularly in subjects with prediabetes.[7]
- Gelesis200 is an additional hydrogel in the Gelesis product family that has a higher elastic modulus and a faster hydration rate. Gelesis200 is being developed for weight loss and glycemic control, particularly in subjects with prediabetes and type 2 diabetes.
- Once ingested, individual Gelesis200 particles hydrate in the stomach and mix homogenously with food, enhancing the volume, elasticity, and viscosity of the stomach and small intestine contents **(Figure 1)**.
- **The purpose of this analysis of the STAGE (Safety, Timing, Appetite and Glycemic Effects) first-in-human study was to determine the effect of Gelesis200 on subjective appetite feelings following 2 or 3 administrations in a single day**.

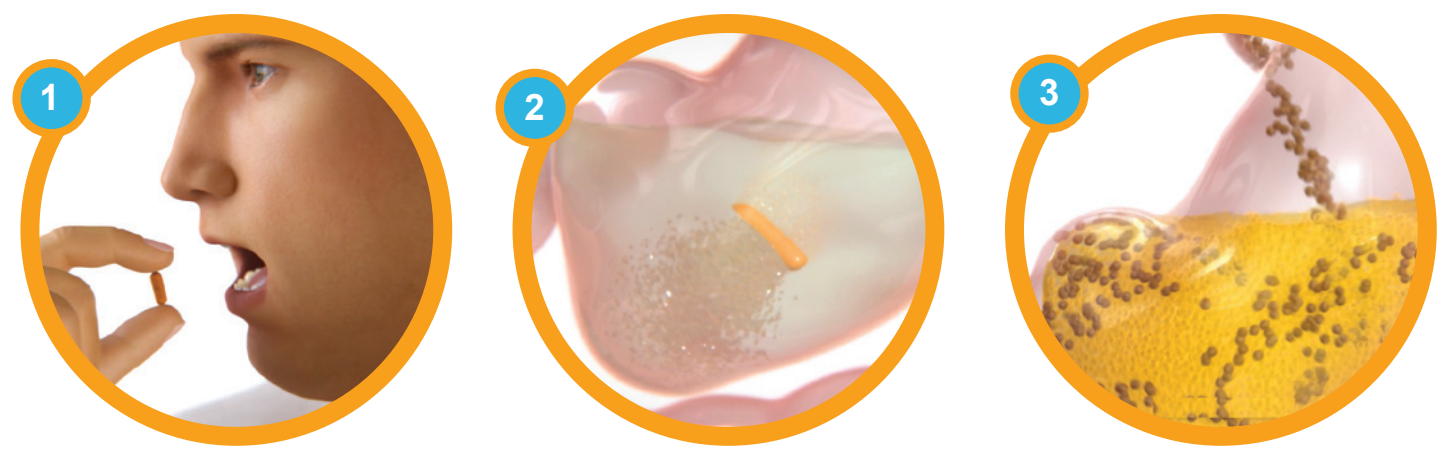
1
2
3
Gelesis capsules are administered with water prior to a meal
Particles release in the stomach and absorb water
Hydrated particles mix homogeneously with food

Ingestion of Gelesis Capsules, and Hydration of Particles

SUBJECTS

- 24 healthy males with BMI 27-35 kg/m^2 and fasting plasma glucose between 90 and <126 mg/dL were enrolled.

CONCLUSIONS

- Short-term Gelesis200 administration in this first-in-human study was safe and well tolerated.
- Administration of Gelesis200 10 min before meals increased subjective feelings of fullness and satiety which is consistent with faster hydration kinetics.
- Taken together, these data support further studies on chronic administration of Gelesis200, a rapidly acting hydrogel, in the management of obesity.

REFERENCES

1. World Health Organization. Obesity and Overweight Fact Sheet. Updated June 2016. http://www.who.int/mediacentre/factsheets/fs311/en/. Accessed October 17, 2016.
2. NCD Risk Factor Collaboration. Worldwide trends in diabetes since 1980: a pooled analysis of 751 population-based studies with 4.4 million participants. *The Lancet*, 2016, 387, 1513-1530.
3. National Diabetes Statistics Report, 2014. http://professional.diabetes.org/04. Goldstein DJ. Beneficial health effects of modest weight loss. *Int J Obes*, 1992, 16, 397-415.
5. Thomas CE et al. Low adoption of weight loss medications: a comparison of prescribing patterns of antiobesity pharmacotherapies and SGLT2s. *Obesity*, 2016, 24, 1955-1961.
6. Heshmati HM et al. Attiva, a novel superabsorbent biodegradable hydrogel, increases the feeling of satiety in humans. In: Program of the 19th Annual Meeting and Clinical Congress of the American Association of Clinical Endocrinologists. April 21-25, 2010; Boston MA.
7. Astrup A et al. Oral administration of Gelesis100, a novel hydrogel, significantly decreases body weight in overweight and obese subjects. In: Program of the Endocrine Society 96th Annual Meeting. June 21-24, 2014; Chicago, IL.

Poster presented at Obesity Week 2016,
October 31 - November 4, New Orleans, LA
Poster number: T-P-LB-3652

METHODS

- This was a single-center, double-blind, placebo-controlled, 4-arm crossover study.
- Subjects were randomly assigned to one of two cohorts as shown in **Figure 2**:
 - **Cohort 1 (BID):** Gelesis 200 (2.10 g total) before breakfast and lunch
 - **Cohort 2 (TID):** Gelesis200 (2.10 g total) before breakfast, lunch, and dinner
- Standard meals containing approximately 570, 830, and 1200 kcals were administered for breakfast, lunch, and dinner, respectively. The distribution of energy from macronutrients for all meals was 45% carbohydrate, 30% fat, and 25% protein.
- Placebo capsules contained approximately 0.57 g of a mixture of microcrystalline cellulose and maltodextrin in a ratio of approximately 50 ± 10%.
- Subjects completed study arms in a block-randomized sequence, with a washout of 7 days between each arm.
- Appetite parameters were assessed using continuous, 10-cm length visual analog scales (VAS) anchored at each extreme. The VAS were self-administered at approximately -30, -10, 0, 30, 60, 90, 120, 150, 180 and 210 min relative to breakfast, lunch, and dinner (in Cohort 2 subjects).
- 24 healthy males with BMI 27-35 kg/m^2 and fasting plasma glucose between 90 and <126 mg/dL were enrolled. was calculated using the linear trapezoidal method on base-line corrected data. Baseline was defined in one of three ways:
- Incremental area under the curve (iAUC) was calculated using the linear trapezoidal method on base-line corrected data. Baseline was defined in one of three ways:
 - 10 min before breakfast for comparisons where treatment was administered 10 min prior to meals (arms A and C)
 - 30 min before breakfast for comparisons where treatment was administered 30 min prior to meals (arms B and D)
 - 0 min before breakfast for comparisons where treatment was administered 30 min and 10 min prior to meals (arms A and B)
- Scheduled times were used in the calculation of iAUC.
- Data from cohorts 1 and 2 were pooled for breakfast and lunch.
- Analysis of variance (ANOVA) was performed on untransformed iAUC values and individual timepoints at the alpha level of 0.05. Factors incorporated in the models included: Sequence, Subject(Sequence), and Treatment. Sequence was tested using Subject(Sequence) as the error term.
- Comparisons were made between Placebo and Gelesis200 specific to each timing of administration and also between both Gelesis200 arms. Comparisons were performed using ANOVA for each timepoint, meal iAUC and whole day iAUC.

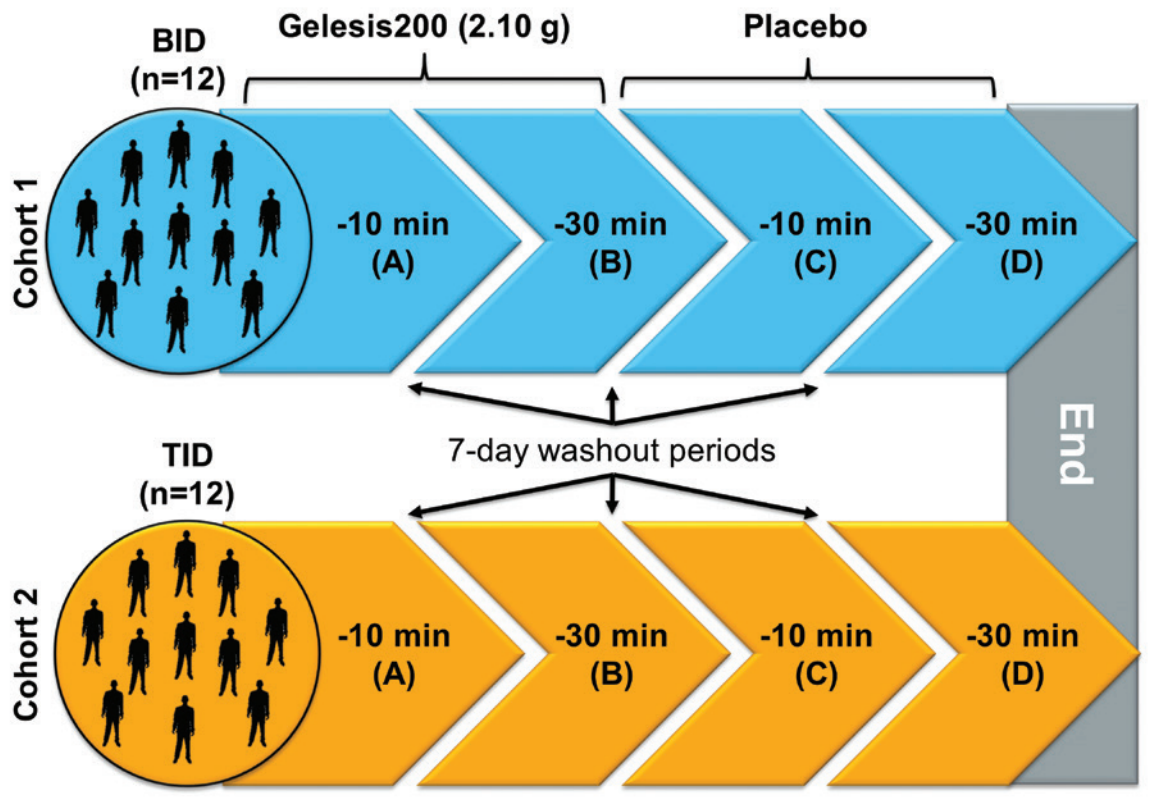

BID
(n=12)
Gelesis200 (2.10 g)
Placebo
Cohort 1
-10 min (A)
-30 min (B)
-10 min (C)
-30 min (D)
7-day washout periods
TID
(n=12)
Cohort 2
-10 min (A)
-30 min (B)
-10 min (C)
-30 min (D)
End

Figure 2: STAGE Study Design. Subjects were randomly assigned to one of two cohorts, and completed Gelesis200 (A: -10 min and B: -30 min prior to meals) and Placebo (C:-10 min and B: -30 min prior to meals) treatment arms in a randomized order with a washout of 7 days between each arm.

RESULTS

- Demographic characteristics of the 24 study subjects who were overweight and obese are provided by cohort in **Table 1**.

Table 1: Subject Demographic Characteristics at Baseline [mean ± SD or N (%)].

	Cohort 1: BID	Cohort 2: TID
N	12	12
Age (y)	42 ± 11	47 ± 7
Body Weight (kg)	89 ± 9	92 ± 5
BMI (kg/m^2)	30 ± 2	30 ± 2
Gender		
Male	12 (100%)	12 (100%)
Race		
White/Caucasian	11 (92%)	10 (83%)
Black	1 (8%)	2 (17%)
Ethnicity		
Hispanic/Latino	0 (0%)	1 (8%)

SAFETY AND TOLERABILITY

- Gelesis200 was safe and well tolerated. The total number of adverse events (AEs) was similar between Gelesis200 and Placebo **(Table 2)**. Nearly all AEs were mild in intensity **(Table 3)**, and no AEs were regarded as being probably related to treatment **(Table 4)**. There were no serious AEs.
- The most common AEs were headache (9), hot flush (5), somnolence (5), procedural dizziness (4).
- Because ingested Gelesis200 is non-systemic and acts entirely within (and is ultimately excreted by) the gastrointestinal (GI) system, GI AEs were examined separately.

Table 2: GI and Other AEs by Treatment Arm and Pooled.

	Treatment Arm					
	Gelesis200		Placebo			
	-10 min (n=24)	-30 min (n=24)	-10 min (n=23)	-30 min (n=21)	Total Gelesis200	Total Placebo
Abdominal discomfort				1		1
Abdominal distension	1			2	1	2
Abdominal pain		1			1	
Eructation			1	1		2
Feces pale		1		1	1	1
Feces soft		1			1	
Flatulence				2		2
Gastroesophageal reflux disease				1		1
Nausea			2			2
All GI AEs	1	3	3	8	4	11
All Other AEs	9	11	9	7	20	16
TOTAL	**10**	**14**	**12**	**15**	**24**	**27**

Table 3: AE Intensity by Treatment Arm and Pooled.

	Treatment Arm					
	Gelesis200		Placebo			
AE Intensity	-10 min (n=24)	-30 min (n=24)	-10 min (n=23)	-30 min (n=21)	Total Gelesis200	Total Placebo
Mild	9	14	10	15	23	25
Moderate	1	0	2	0	1	2
Severe	0	0	0	0	0	0

Table 4: AE Causality by Treatment Arm and Pooled.

	Treatment Arm					
	Gelesis200		Placebo			
AE Causality	-10 min (n=24)	-30 min (n=24)	-10 min (n=23)	-30 min (n=21)	Total Gelesis200	Total Placebo
Unrelated	7	2	1	3	9	4
Remote	0	1	2	0	1	2
Possibly	3	11	9	12	14	21
Probably	0	0	0	0	0	0

EFFECTS OF GELESIS200 ON APPETITE

- Compared to Placebo, administration of Gelesis200 10 min prior to meals resulted in greater fullness after lunch **(Figure 3A)**. The 12 subjects who completed all 3 meals reported greater fullness during the entire day **(Table 5)**.
- Administration of Gelesis200 10 min prior to meals resulted in greater satiety just before dinner, and 150 and 180 min after dinner **(Figure 3B)**. A trend (P = 0.075) was observed for greater satiety during the entire day in the 12 subjects who completed all 3 meals **(Table 5)**.
- When Gelesis200 was administered 30 min prior to meals, the above differences in fullness and satiety were not observed.
- Compared to Gelesis200 administration 30 min prior to meals, Gelesis200 administration at 10 min prior to meals produced more fullness 90 min after lunch (1.0 ± 2.5 cm; P = 0.050) and less prospective food intake 30 min (-0.8 ± 1.7 cm; P = 0.037), 90 min (-1.3 ± 2.0 cm; P = 0.005), and 120 min (-1.0 ± 2.1 cm; P = 0.039) after lunch.

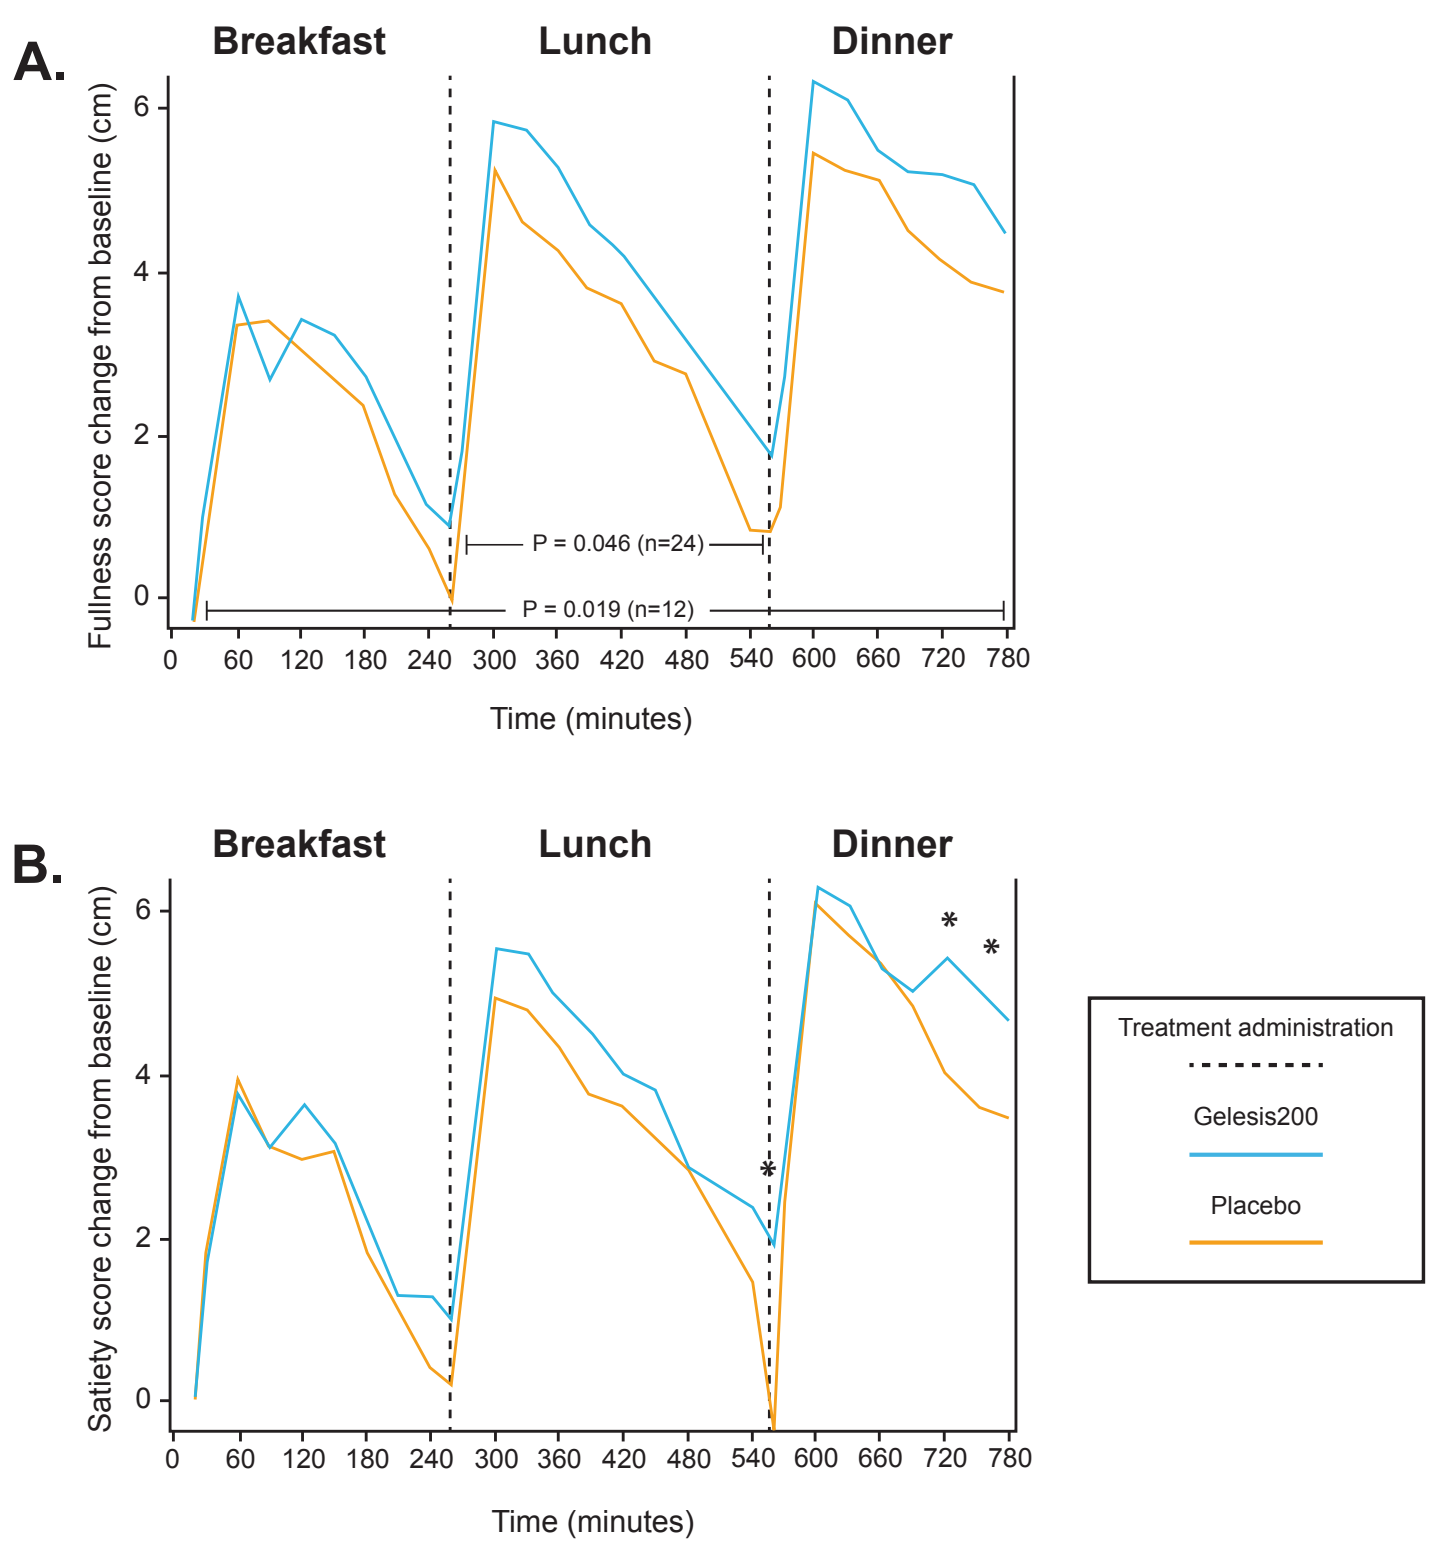

A.
Breakfast
Lunch
Dinner
Fullness score change from baseline (cm)
P = 0.046 (n=24)
P = 0.019 (n=12)
Time (minutes)
B.
Breakfast
Lunch
Dinner
Satiety score change from baseline (cm)
*
Time (minutes)
Treatment administration
Gelesis200
Placebo

Figure 3: Changes in Fullness (Panel A) and Satiety (Panel B) Following Administration of Gelesis200 or Placebo 10 Min Prior to Meals. *P = 0.017, 0.032, and 0.031 at 10 Min Prior to Dinner, and 150 and 180 Min After Dinner, Respectively.

Table 5: Incremental AUC for Fullness and Satiety When Gelesis200 or Placebo Was Administered 10 Min Prior to Meals.

		Fullness iAUC (Mean ± SD)		
	N	Gelesis200	Placebo	*P* values
Breakfast	24, 23	623 ± 451	578 ± 474	0.525
Lunch	24, 23	1053 ± 532	853 ± 576	0.046
Dinner	12	1234 ± 682	1019 ± 660	0.086
Total Day	**12**	2963 ± 2007	2129 ± 1689	0.019
		Satiety iAUC (Mean ± SD)		***P* values**
	N	**Gelesis200**	**Placebo**	
Breakfast	24, 23	561 ± 509	520 ± 356	0.700
Lunch	24, 23	956 ± 601	810 ± 549	0.169
Dinner	12	1173 ± 706	993 ± 557	0.096
Total Day	**12**	2867 ± 2126	2215 ± 1593	0.075

DISCUSSION

- Despite well-documented benefits of weight loss on obesity-related comorbidities,[4] adoption of antiobesity pharmacotherapies is lagging[5] likely due to safety concerns based on a history of FDA withdrawals. This highlights an unmet need for safe and effective therapies to facilitate the reduction of excess weight.
- Administration of Gelesis200 10 min prior to meals induced subjective feelings of fullness and satiety. This is consistent with previously reported appetite effects of Gelesis100.[6] However, administration of Gelesis200 at 10 min prior to lunch was better at producing feelings of fullness and reducing prospective food intake (i.e., desire to eat) than administration 30 min prior to lunch which is consistent with the faster hydration kinetics of Gelesis200.
- Gelesis200, is similar in concept to Gelesis100, but has different physical properties that could potentially address different indications and treatment regimens. Gelesis200 hydrates more rapidly than Gelesis100 and creates a higher elastic response and viscosity, but occupies a slightly smaller volume in the stomach. For example, due to its higher elastic response and accelerated hydration, Gelesis200 could be more suitable for glycemic control, where one relevant mechanism is the delay of the absorption of glucose in the small intestine. For this purpose, the volumetric effect at the beginning of the meal could be less important and Gelesis200 could be administered immediately prior to the meal. These properties could make Gelesis200 more suitable for glycemic control in subjects with prediabetes and type 2 diabetes, who may or may not require weight loss.
- In this study, Gelesis200 was safe and well tolerated. Overall, AEs were mild in intensity, were similar in frequency between Gelesis200 and Placebo treatment arms, and no AEs were regarded as being probably related to treatment.
- Given the overall benefit-risk profile observed in this first-in-human study, further studies on chronic administration could be promising.